The 14th WRIB

Pharmacodyanamic Analysis of Crizanlizumab by Surface Plasmon Resonance: A Case for Implementing Unique Analytical Platforms in Clinical Trials

Shuo Wang[1], David Escobar[1], Sergei Agoulnik[2], Violet Abraham[1], Anil Pahuja[1], James Zhou[1], David Floch[3], Mikiko Iwata[1], Manuel Cajina[2], Lauren Nardini[2], Ron Linnartz[2], Shabnam Tangri[1] and Naveen Dakappagari[1]

[1]Navigate Biopharma Services, Inc., a Novartis subsidiary, Carlsbad, CA; [2]Precision Medicine, Novartis, Cambridge, MA; [3]PK Sciences, Novartis, Basel, Switzerland

BACKGROUND

- ➢ Crizanlizumab is the first and only FDA-approved monoclonal antibody (marketed as Adakveo®) that reduces the frequency of vaso-occlusive crises (VOCs) or pain crises in adults and pediatric patients, aged 16 years and older, with sickle cell disease.
- ➢ This antibody binds to P-selectin on endothelial cells and platelets thereby inhibiting its interaction with P-selectin glycoprotein ligand-1 (PSGL-1) expressed on blood cells. The P-selectin-PSGL-1 interaction promotes sticking of blood cells leading to sickle cell related pain crisis.
- ➢ Surface Plasmon Resonance (SPR) has been used routinely in drug discovery due to its ability to measure molecular interactions in a label free high throughput manner. However, this powerful approach has not been applied in pivotal clinical studies.
- ➢ We describe here the development and validation of an SPR based pharmacodynamics (PD) assay utilized to demonstrate the inhibition of P-selectin-PSGL-1 interaction by crizanlizumab and the key challenges encountered with introduction of this technology into a clinical trial setting.

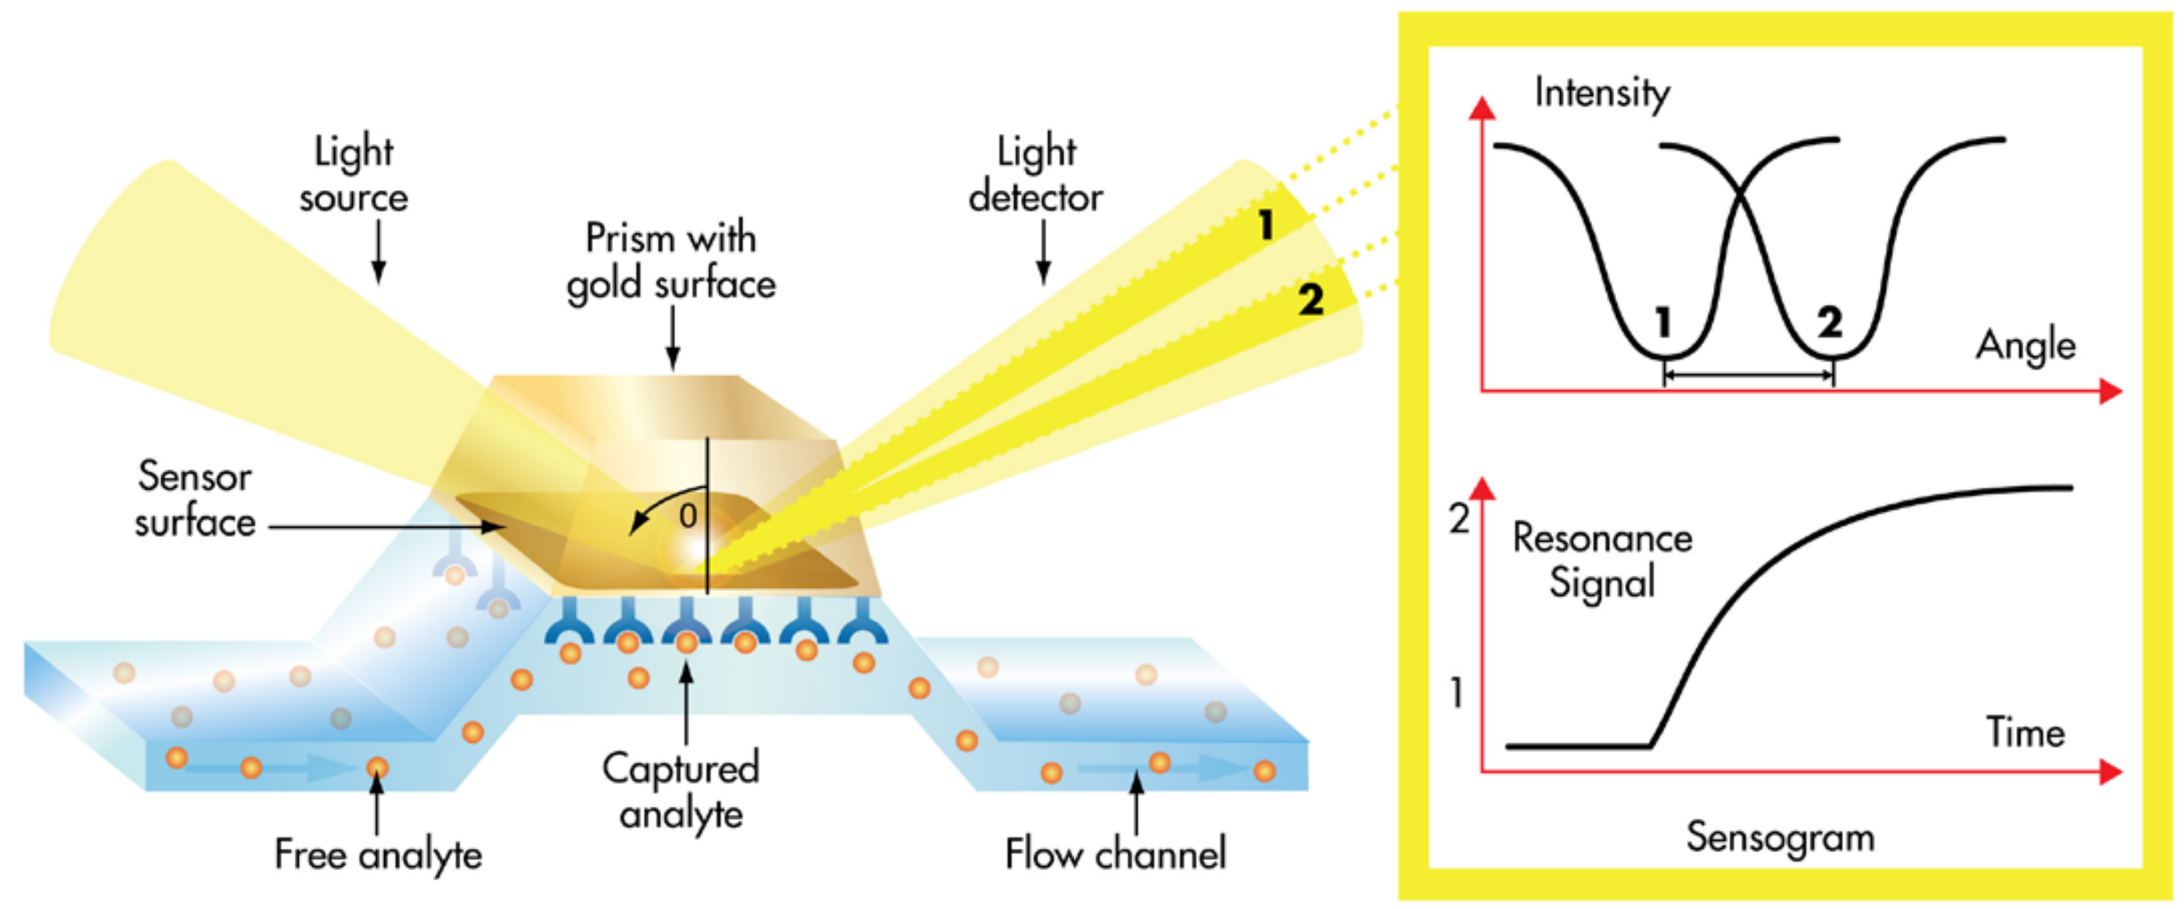

Light source
Prism with gold surface
Light detector
Sensor surface
Captured analyte
Free analyte
Flow channel
Intensity
Angle
Resonance Signal
Time
Sensogram

Figure 1. Schematic configuration of a label-free detection using SPR.
(Adapted from www.bruker.com)

METHODS

- ➢ The SPR assay to quantify inhibitory (PD) activity of crizanlizumab was designed to mimic in vivo biological interactions, wherein, a biotinylated glycosulfopeptide-6 (GSP6) representing the PSGL-1 binding domain was immobilized on a streptavidin coated sensor chip.
- ➢ Serum samples from patients treated with crizanlizumab were injected over the GSP6 sensor chip in the presence of excess P-selectin and inhibitory activity of the drug (function of serum concentration) was measured as change in response units from the baseline sample.

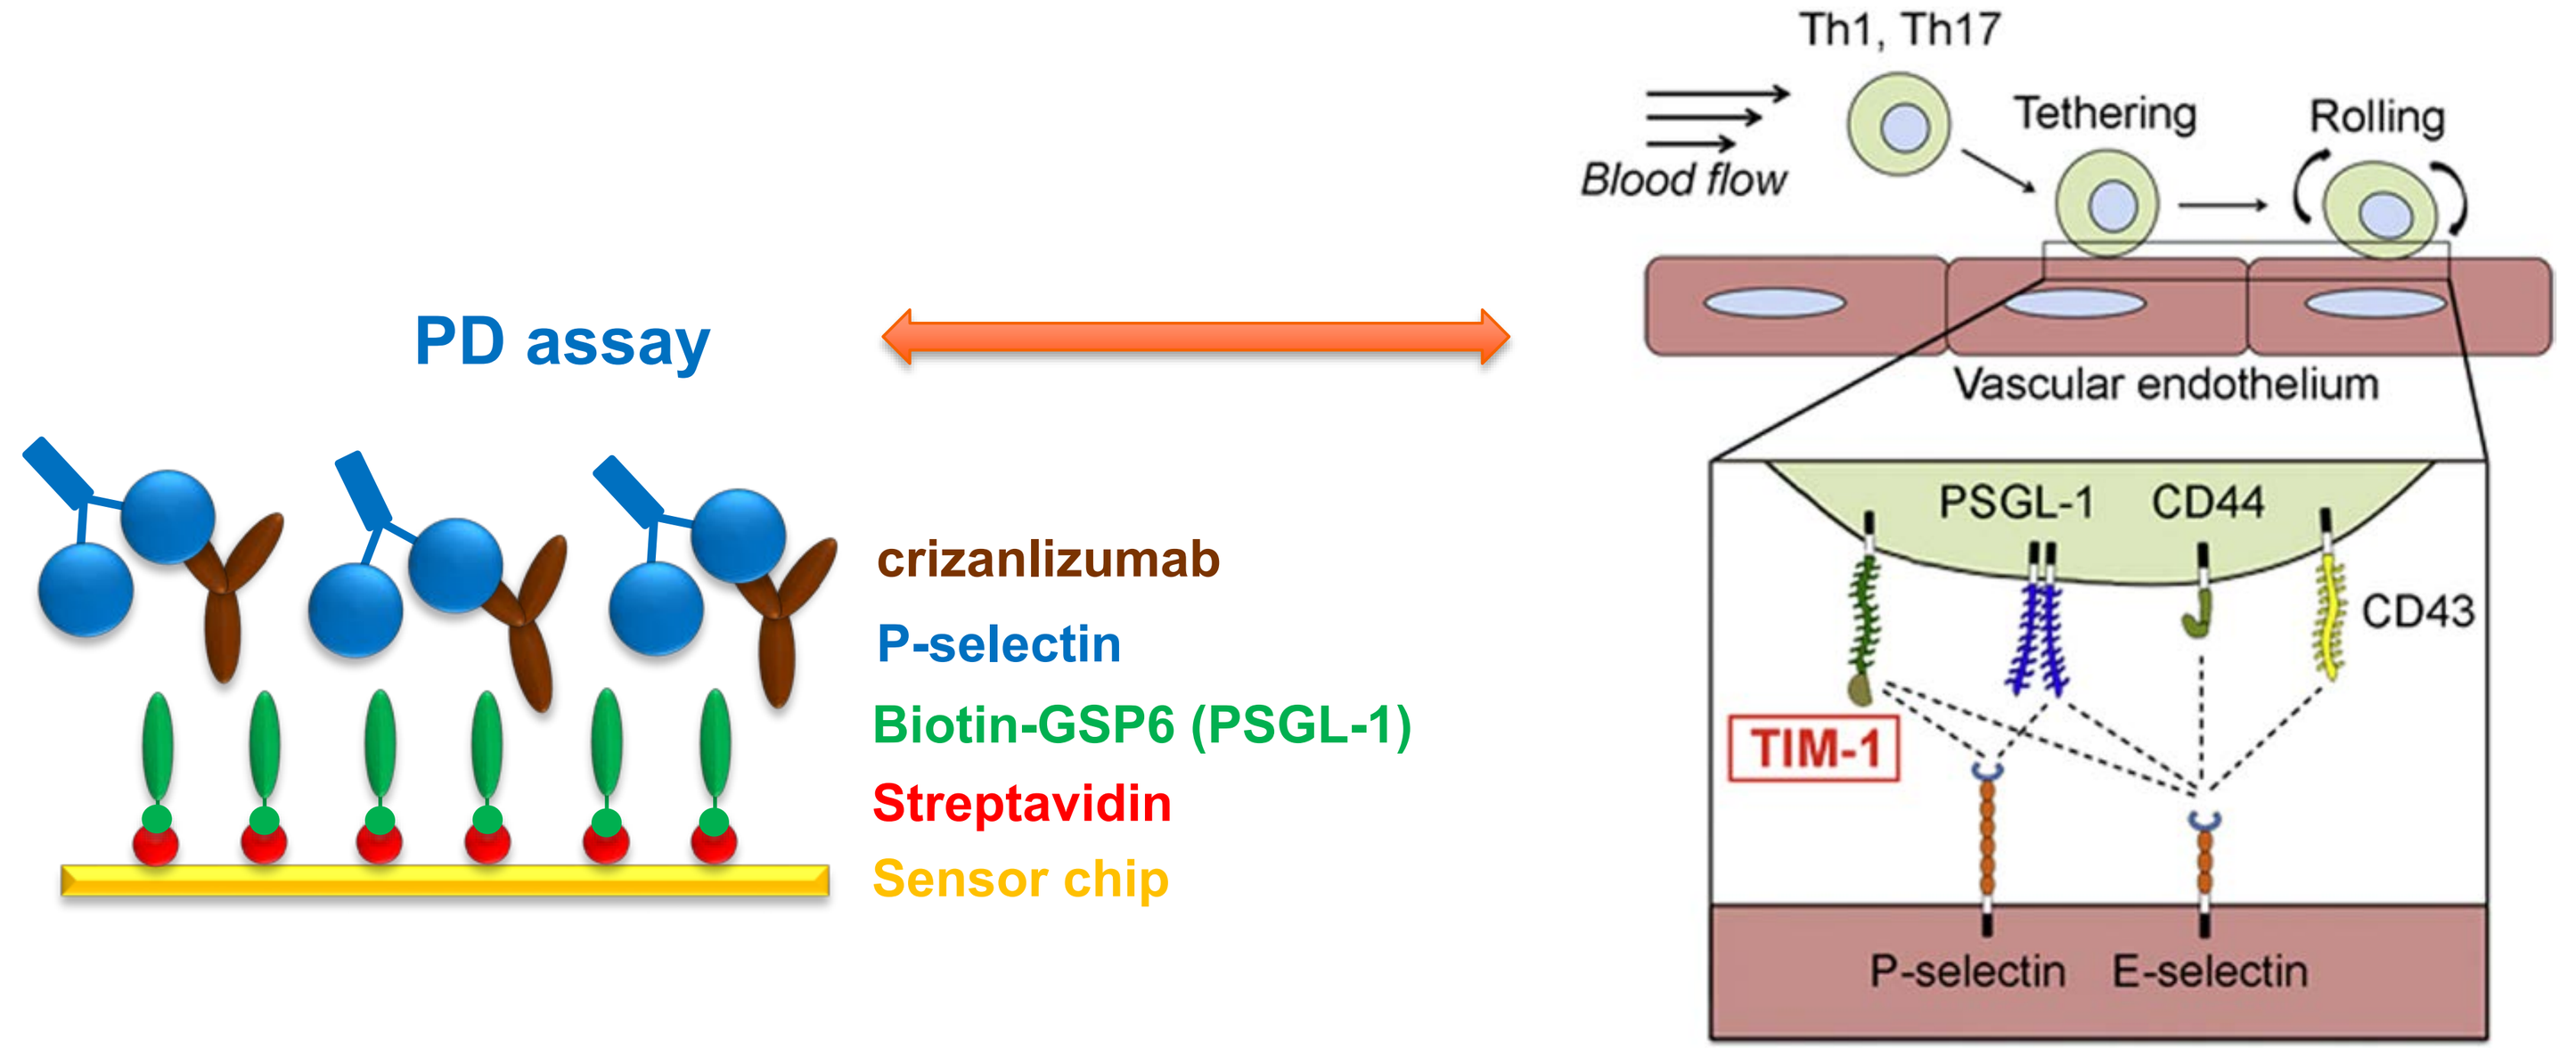

PD assay
crizanlizumab
P-selectin
Biotin-GSP6 (PSGL-1)
Streptavidin
Sensor chip
Th1, Th17
Blood flow
Tethering
Rolling
Vascular endothelium
PSGL-1
CD44
CD43
TIM-1
P-selectin
E-selectin

Figure 2. In vivo biology derived PD assay design principle.
(Right figure is from Immunity 2014, Volume 4 issue 4, page 542-553)

ASSAY DEVELOPMENT

- ➢ Optimize GSP6 immobilization density to improve the longevity of the sensor chip's surface
 - Minimize avidity and P-selectin binding signal loss
 - Include surface conditioning cycles prior to clinical sample testing

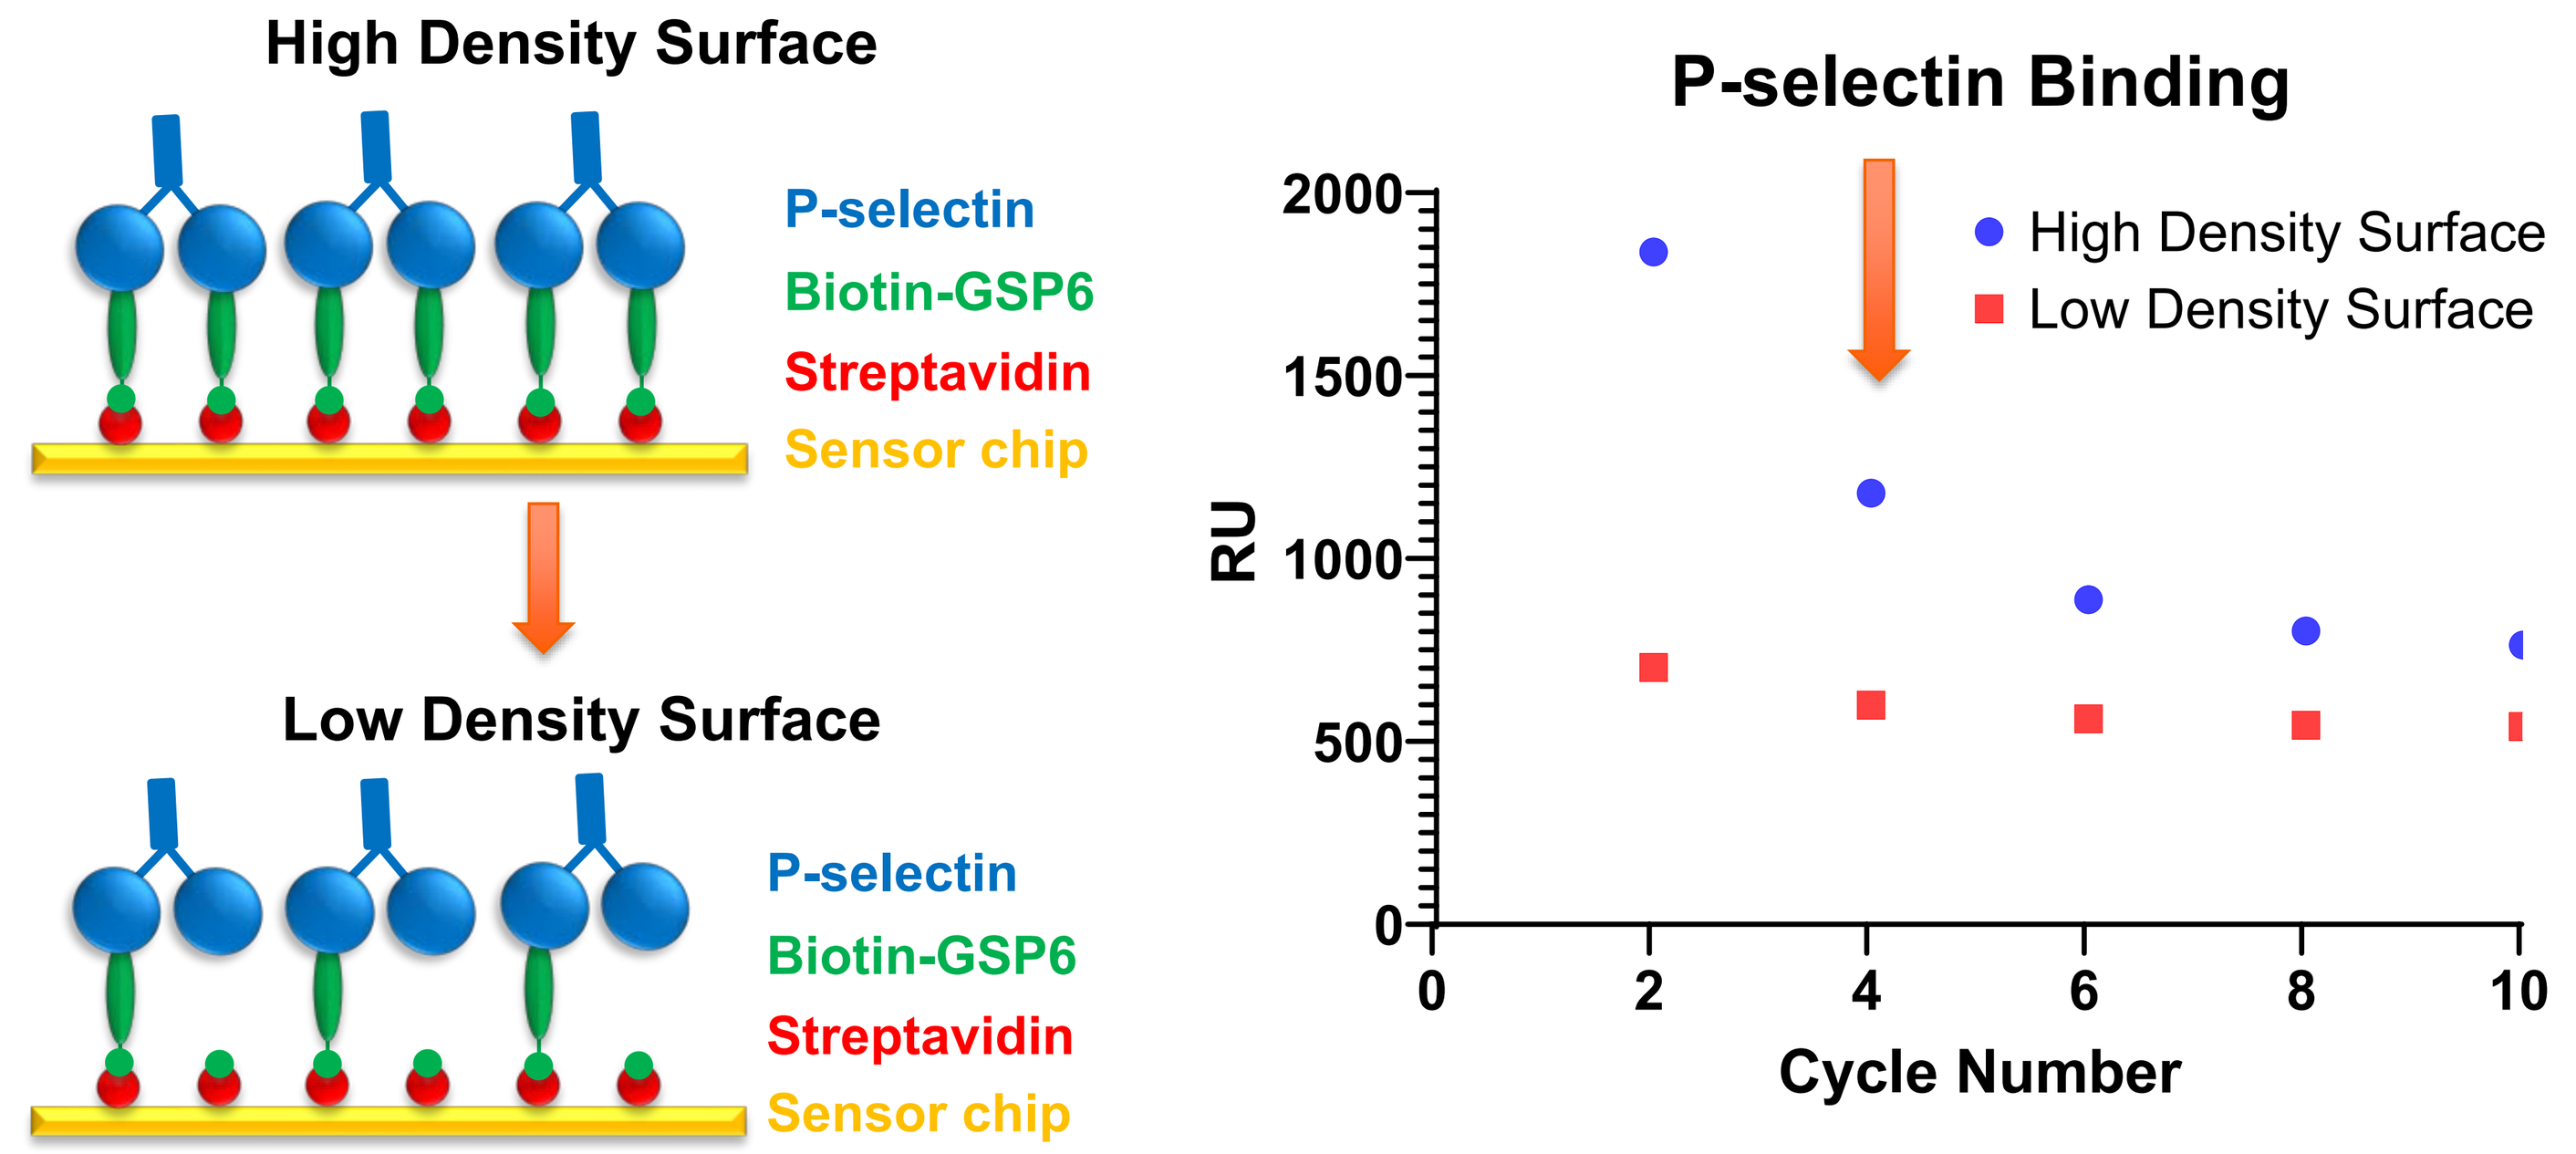

High Density Surface
P-selectin
Biotin-GSP6
Streptavidin
Sensor chip
Low Density Surface
P-selectin
Biotin-GSP6
Streptavidin
Sensor chip
P-selectin Binding
High Density Surface
Low Density Surface
RU
2000
1500
1000
500
0
0
2
4
6
8
10
Cycle Number

Figure 3. Lowering the immobilization density of GSP6 has shown to minimize the signal loss from P-selectin binding and increase longevity of biosensor surface.

ASSAY VALIDATION

- ➢ Standard inhibition curve of crizanlizumab
 - 11 concentrations titrated with a fixed concentration of P-selectin
 - Negative Control (buffer only) and Positive Control (P-selectin only) are included to calculate %binding of P-selectin and %inhibition of crizanlizumab (100% -%binding = %inhibition)
 - IC_{50} value is generated and used as the bench mark to bridge critical reagent changes, such as different lots of P-selectin and GSP6
 - Crizanlizumab shows comparable IC_{50} values in different matrix
 - Three levels of assay quality controls (QCs) are determined: HQC, MQC and LQC

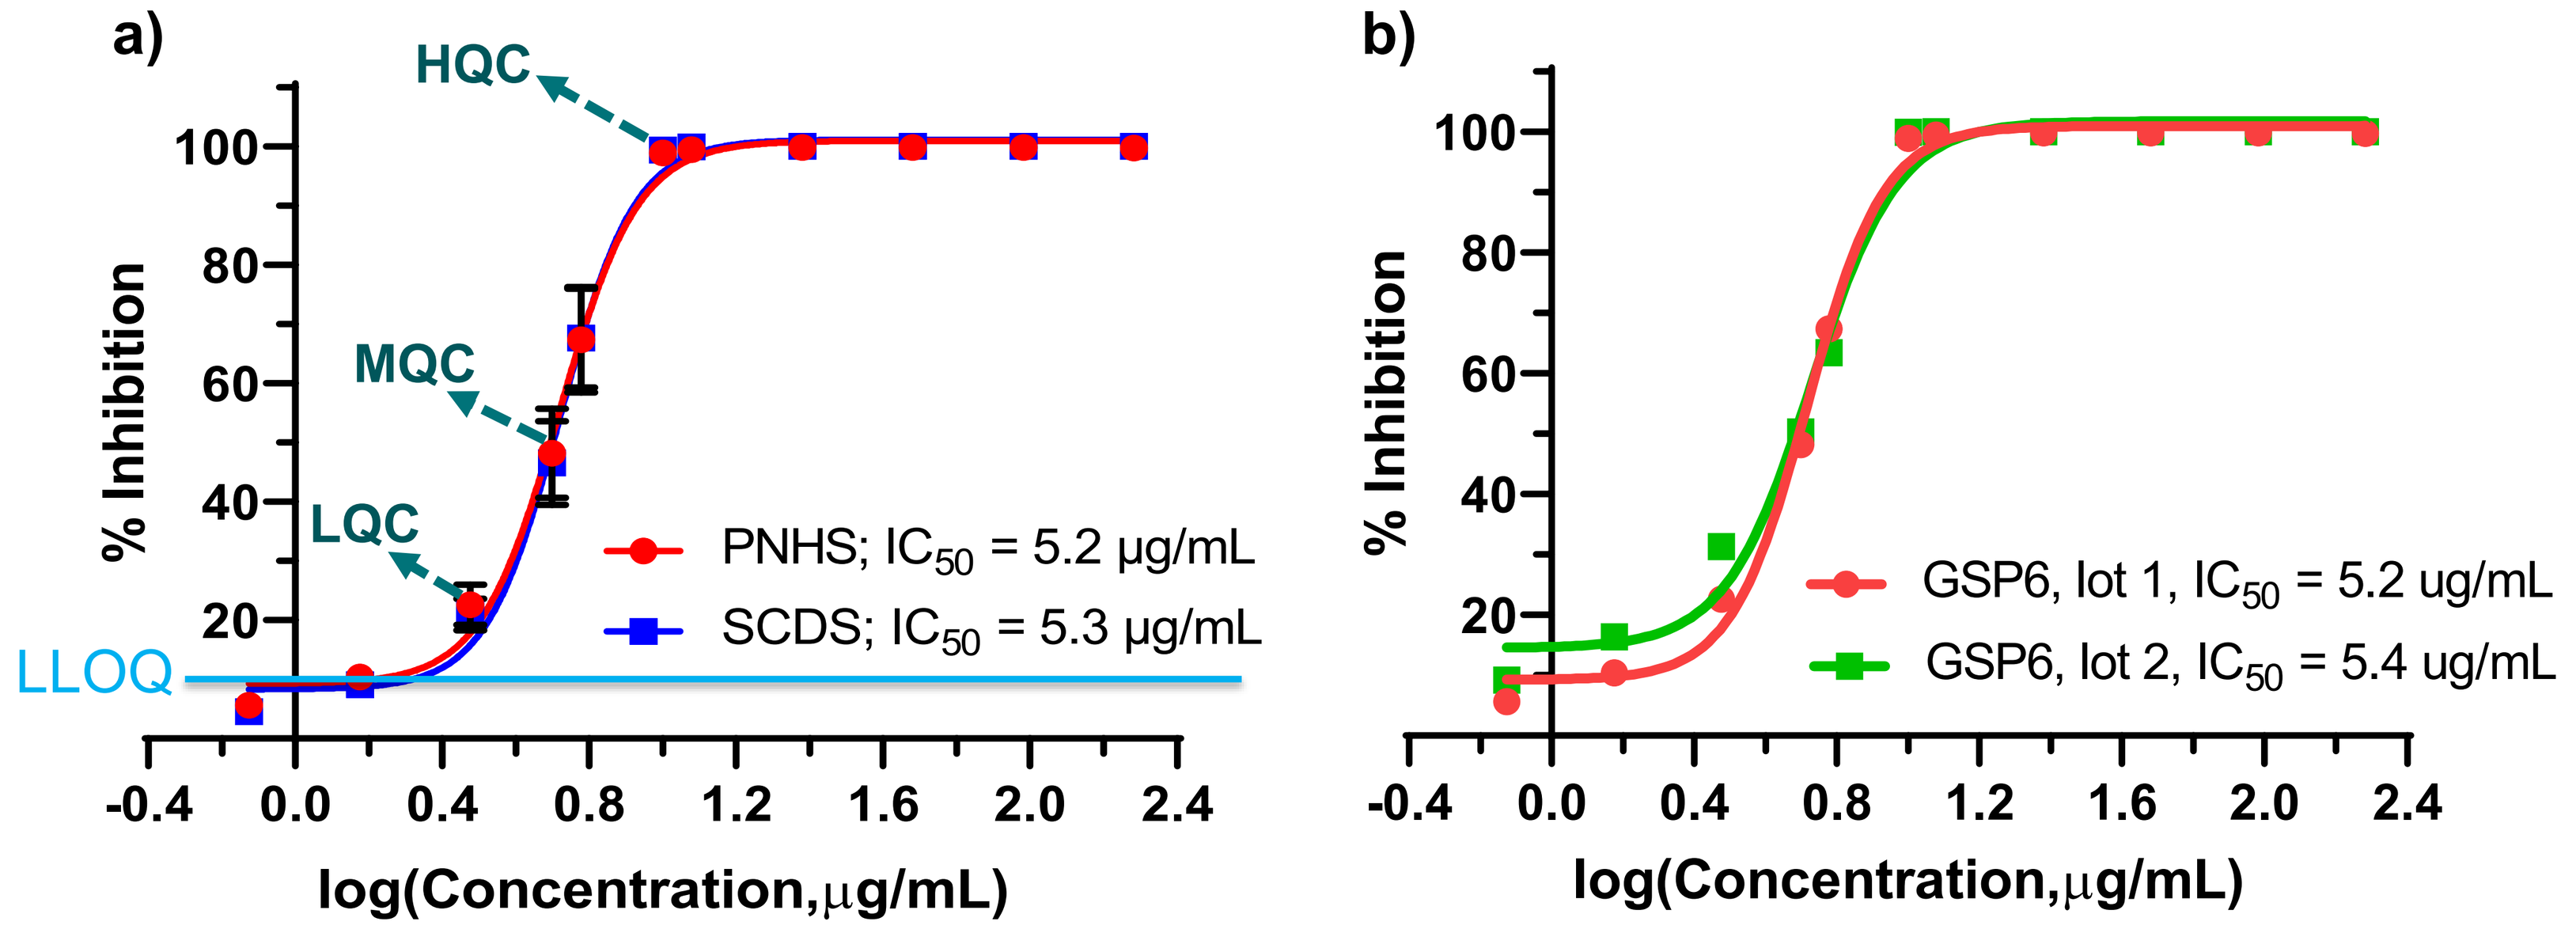

a)
HQC
MQC
LQC
LLOQ
% Inhibition
PNHS; IC50 = 5.2 µg/mL
SCDS; IC50 = 5.3 µg/mL
log(Concentration,µg/mL)
b)
% Inhibition
GSP6, lot 1, IC50 = 5.2 ug/mL
GSP6, lot 2, IC50 = 5.4 ug/mL
log(Concentration,µg/mL)

PNHS = Pooled Normal Human Serum
SCDS = Sickle Cell Disease Serum

Figure 4. a) Crizanlizumab inhibition curve in PNHS and SCDS
b) Crizanlizumab inhibition curve in PNHS with two different lots of GSP6

ANALYTICAL VALIDATION RESULTS SUMMARY

Parameter	Result	Acceptance Criteria	Outcome (P/F)
Inhibition Curve (Linearity of dilution)	Concentration range: 0.75 – 192 µg/mL %CV = 19.2% IC_{50} = 5.2 µg/mL	%CV: ≤ 20% (≤ 25% at LLOQ)	Pass
Total Precision	Highest %CV = 21.9% (at LLOQ)	%CV: ≤ 20% (≤ 25% at LLOQ)	Pass
Accuracy	Highest % Recovery = 20.5% Highest %CV = 10.9%	% RPD within ±25% %CV: ≤ 20% (≤ 25% at LLOQ)	Pass
Reportable Inhibition Range	ULOQ = 100.0% LLOQ = 10.4%	N/A	N/A
Interference	PNHS IC_{50} = 5.2 µg/mL SCDS IC_{50} = 5.3 µg/mL	IC_{50} from SCD ± 20% from PNHS IC_{50}	Pass

ASSAY PERFORMANCE

- ➢ One set of QCs is tested at the beginning of the assay run and another set is tested at the end of the assay run.
- ➢ Clinical sample results are only considered valid when QC meets pre-defined acceptance criteria.
- ➢ The trend of QC results is monitored for each clinical trial to ensure the proper PD assay performance.
- ➢ HQC inhibition near or at 100% with minimum variation and therefore not shown here.

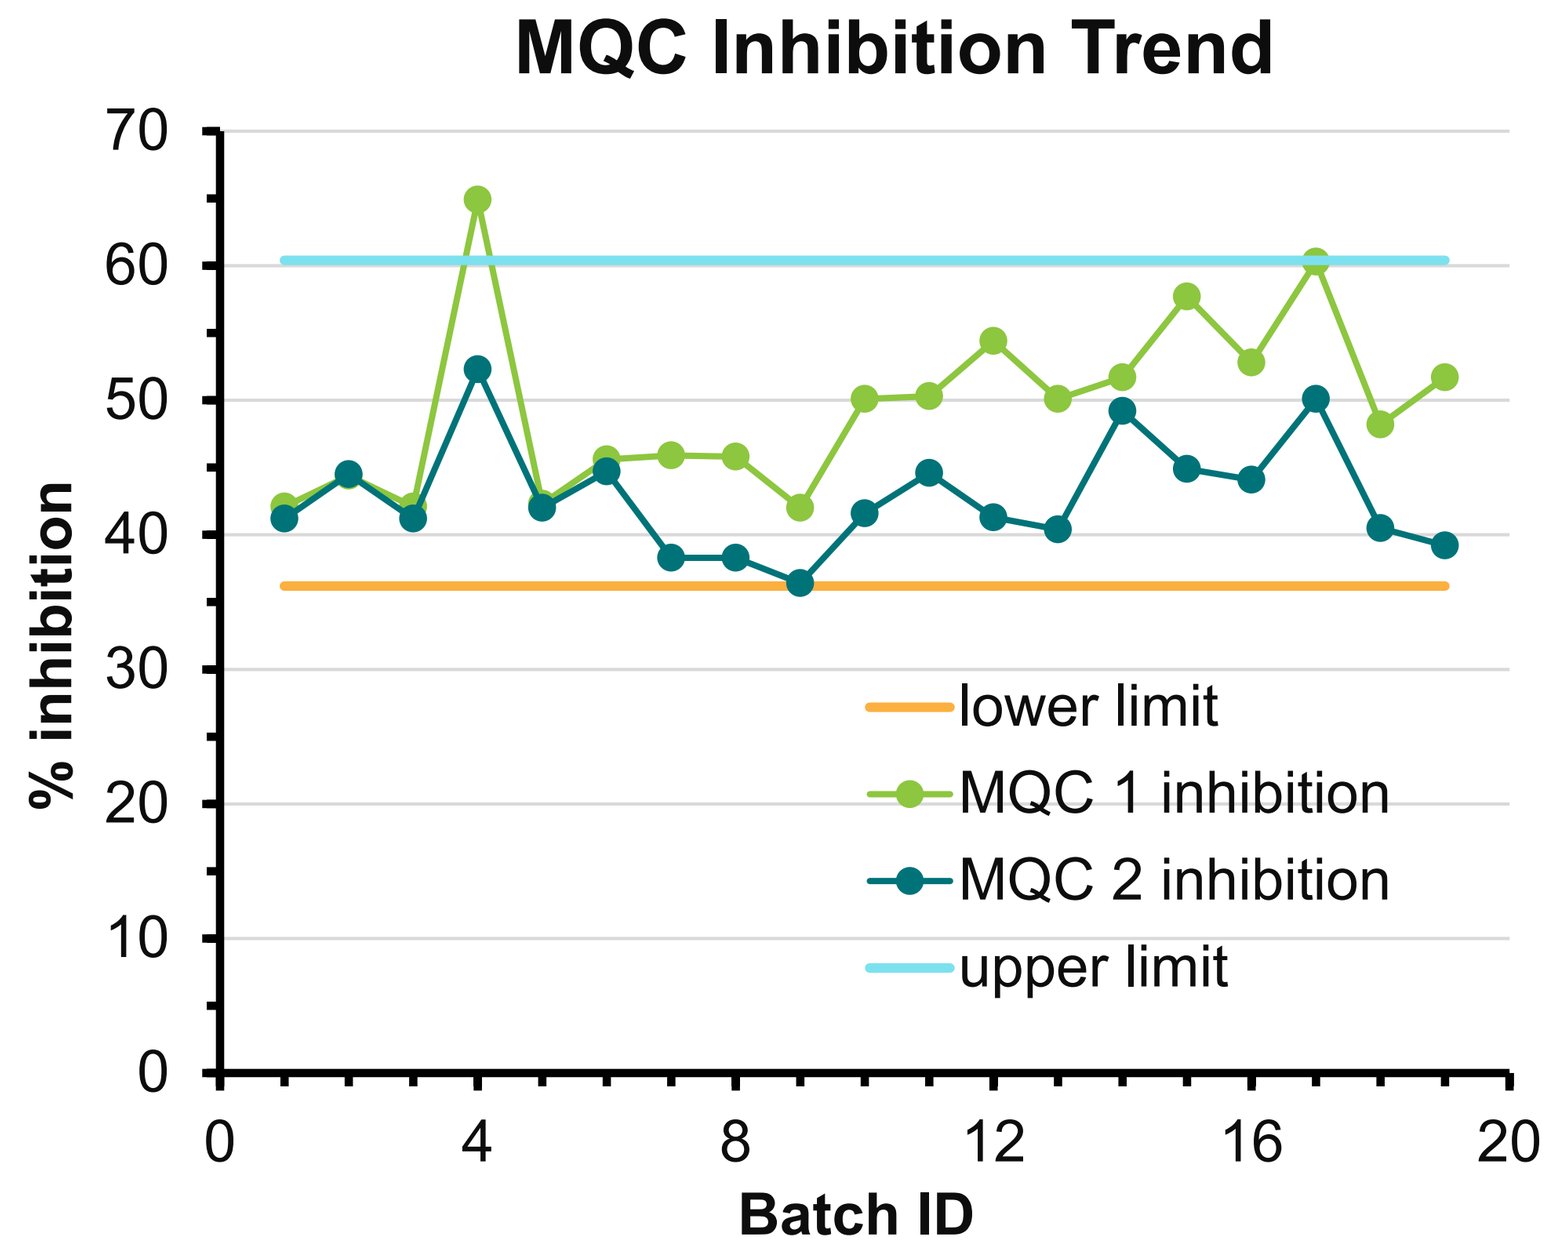

MQC Inhibition Trend
% inhibition
lower limit
MQC 1 inhibition
MQC 2 inhibition
upper limit
Batch ID

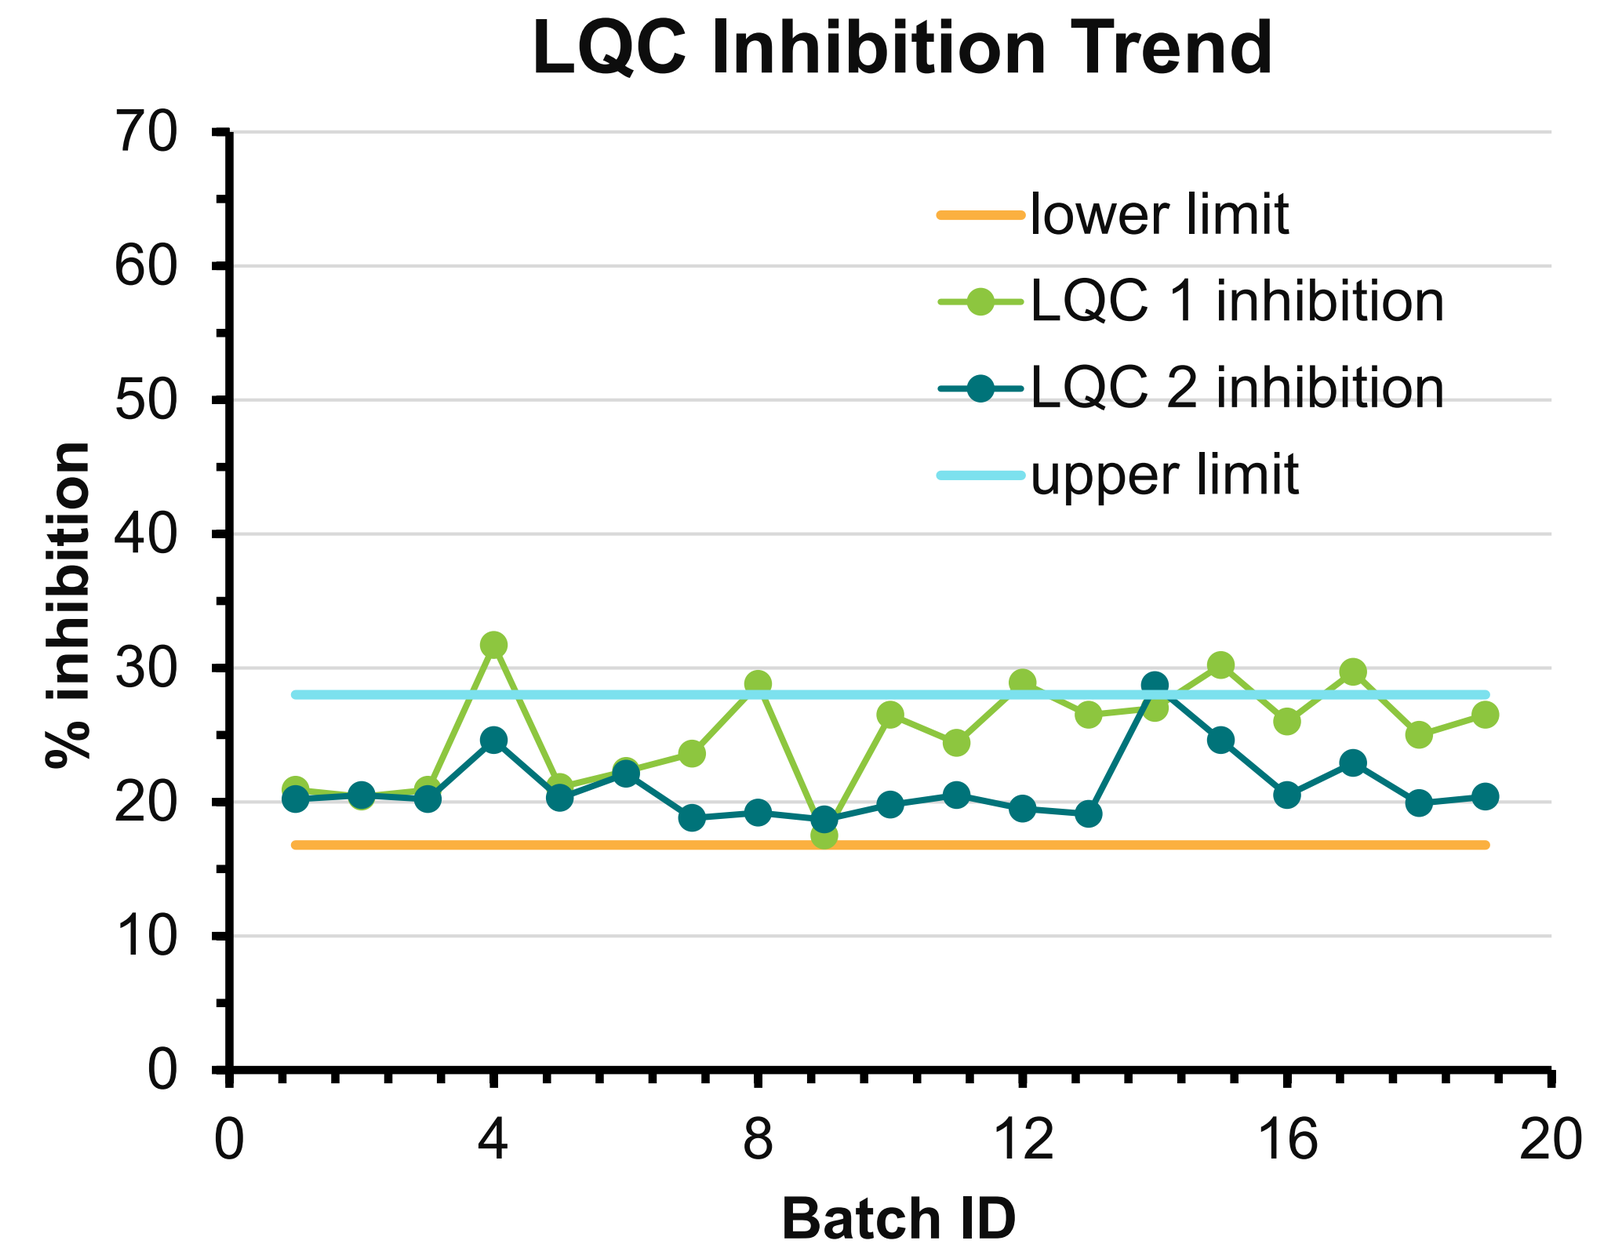

LQC Inhibition Trend
% inhibition
lower limit
LQC 1 inhibition
LQC 2 inhibition
upper limit
Batch ID

PD RESULTS FROM CLINICAL TRIALS

Crizanlizumab showed inhibition of P-selectin binding in clinical studies of sickle cell disease patients, and, the inhibitory properties of crizanlizumab were comparable amongst different manufacturing methods.

Single dose of crizanlizumab with different manufacturing processes in Healthy Volunteers

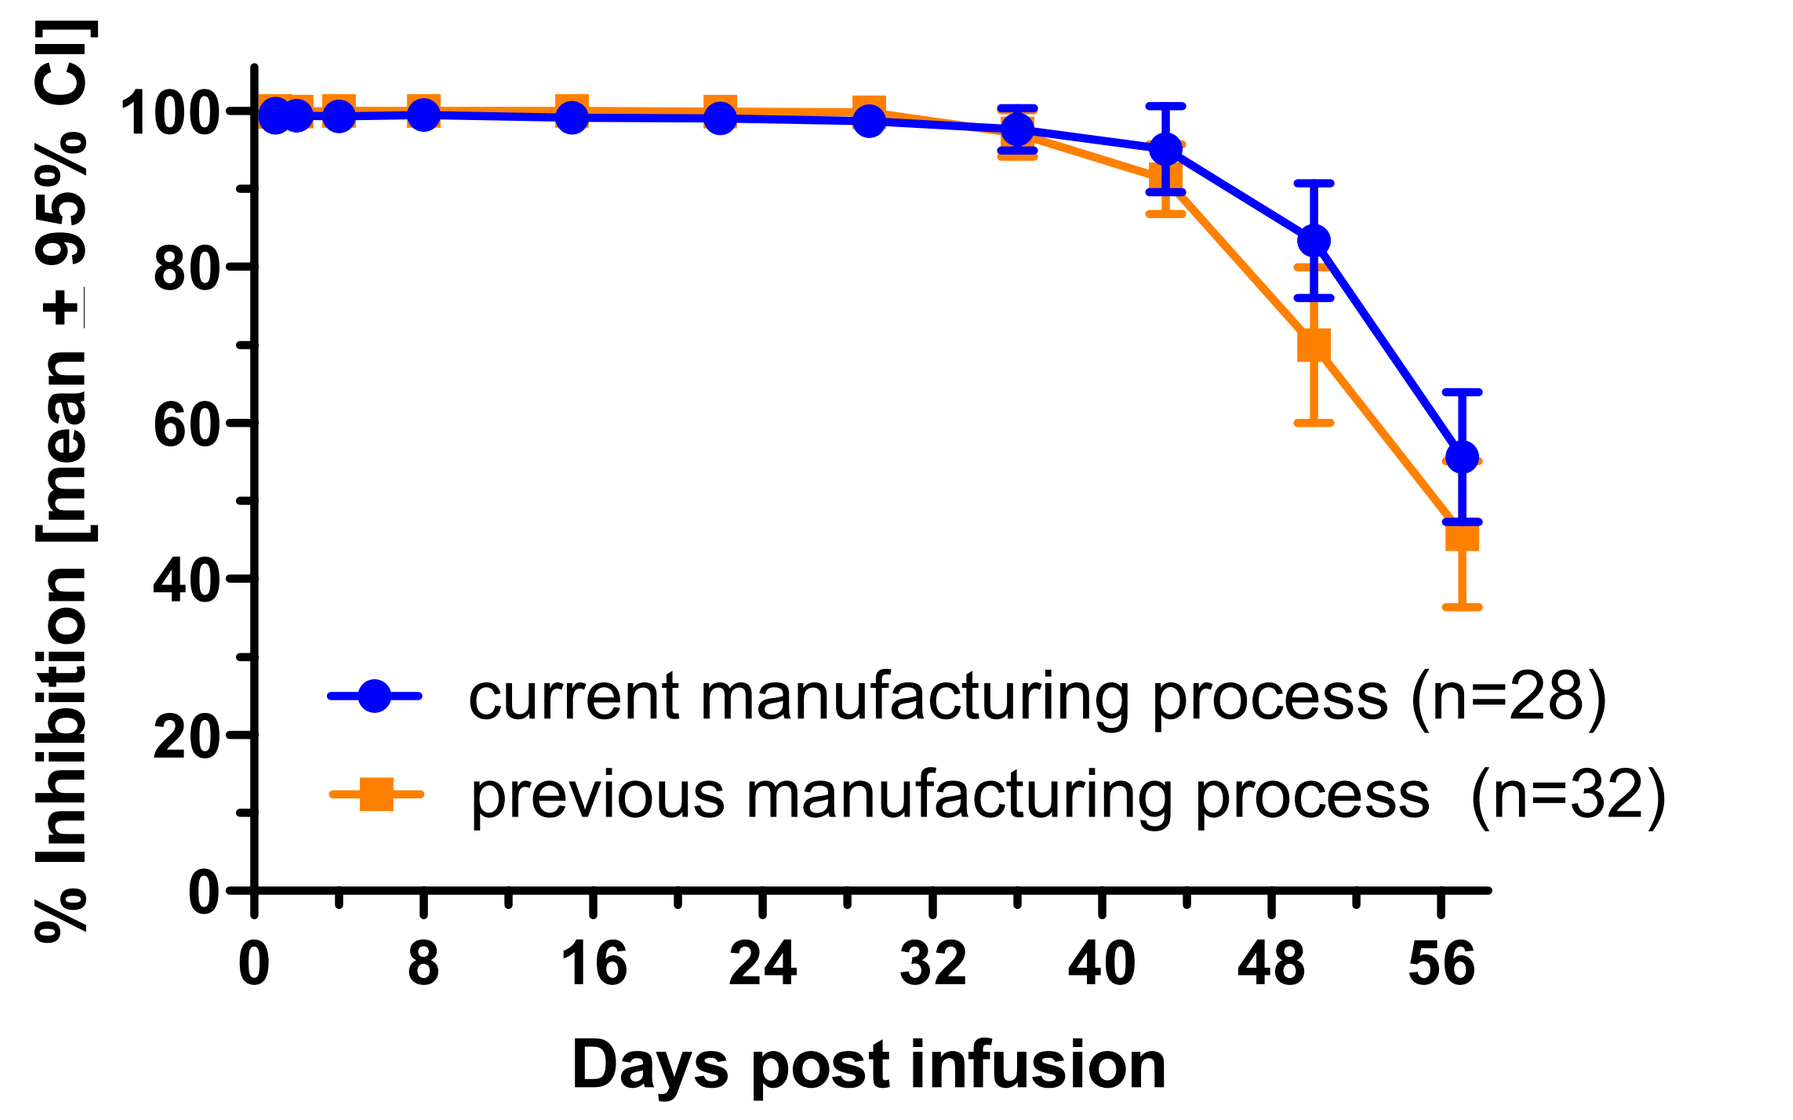

% Inhibition [mean ± 95% CI]
current manufacturing process (n=28)
previous manufacturing process (n=32)
Days post infusion

Multiple dose of crizanlizumab in Sickle Cell Disease Patients*

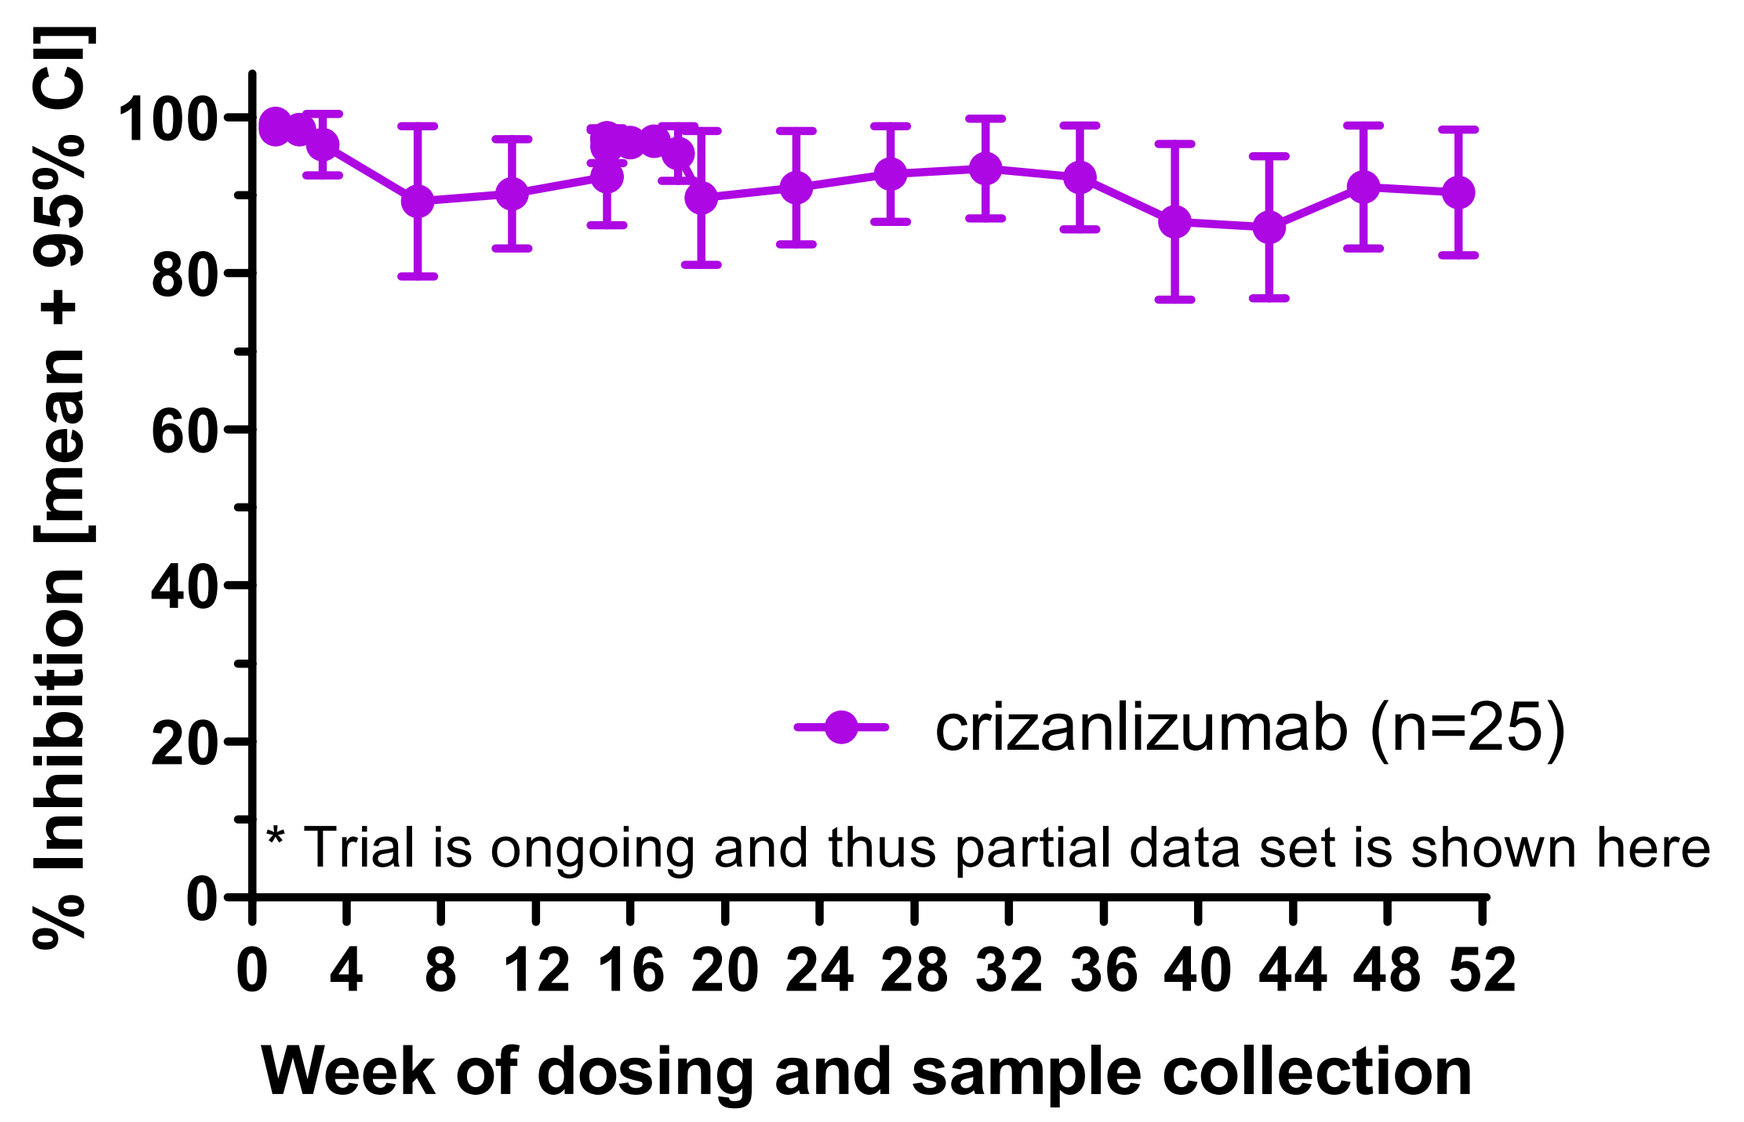

% Inhibition [mean + 95% CI]
crizanlizumab (n=25)
* Trial is ongoing and thus partial data set is shown here
Week of dosing and sample collection

CONCLUSIONS

- ➢ A systematic approach was taken to qualify the instrumentation and novel reagents and perform advanced assay validation commensurate with regulatory guidelines for use of a ligand binding method for a co-primary endpoint of a clinical trial supportive of drug registration.
- ➢ The validated SPR assay has been applied to over 3000 serum samples from clinical trials of crizanlizumab to support a co-primary endpoint and aid in determining the impact of crizanlizumab on P-selectin binding, thus, enabling use of the SPR assay across ongoing sickle cell disease trials.

Scan this QR code

This presentation is the intellectual property of the author/presenter. Contact naveen.dakappagari@navigatebp.com for permission to reprint and/or distribute.